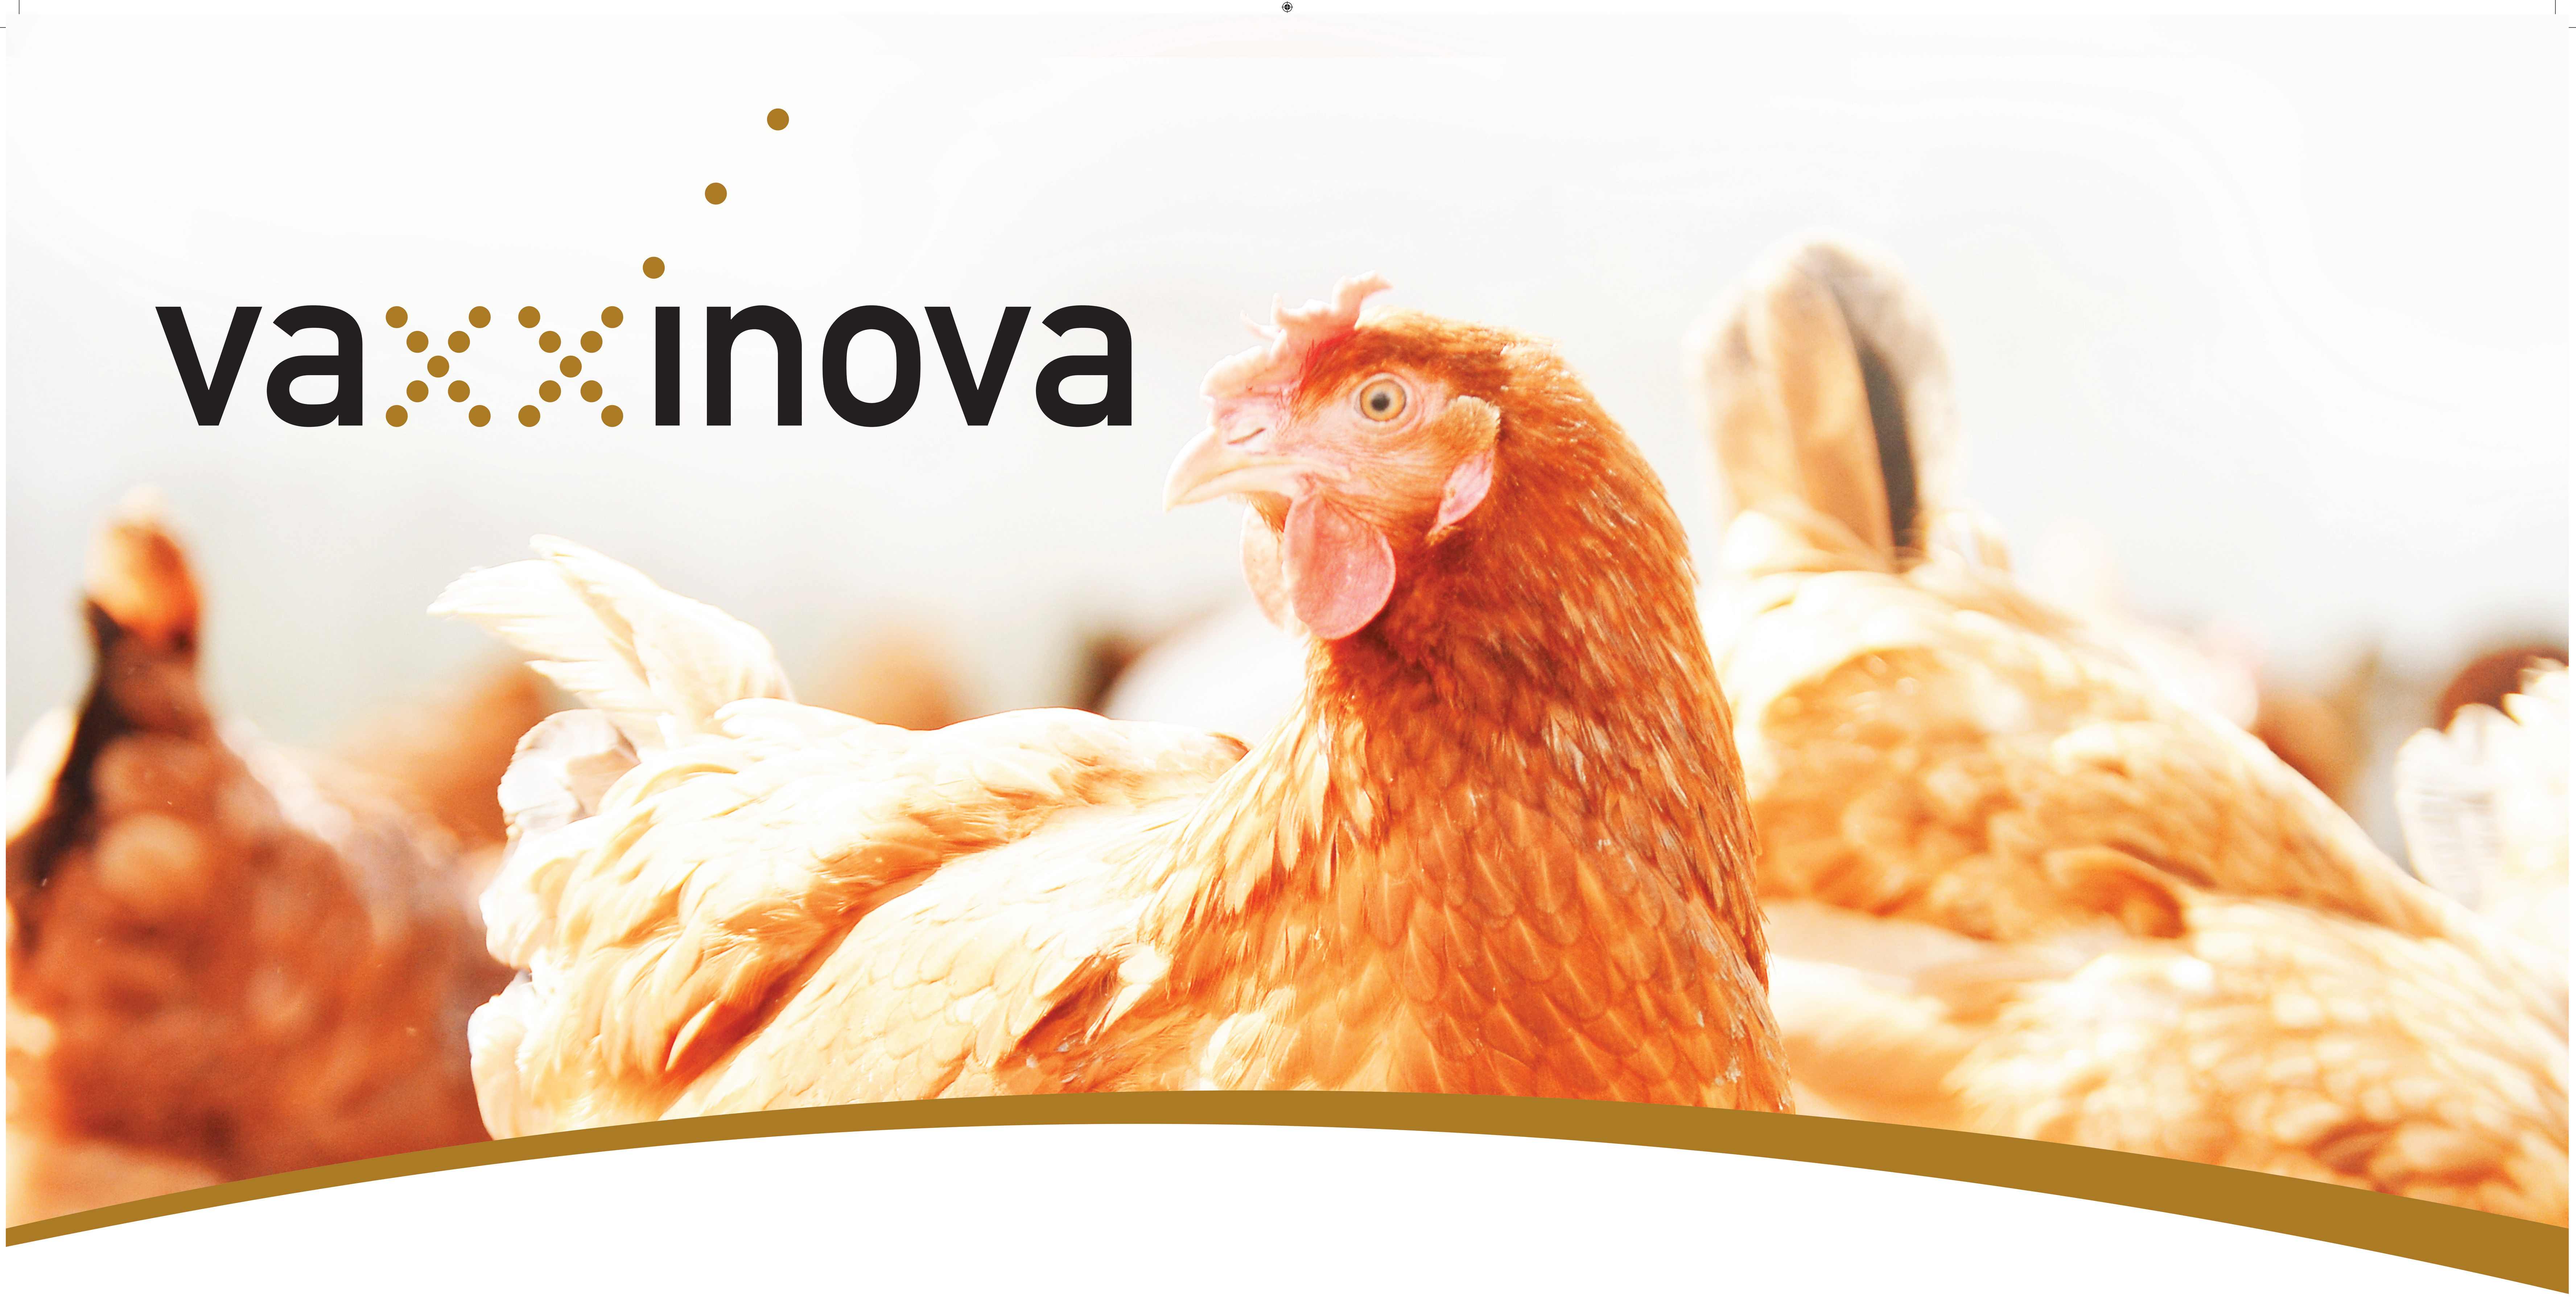
vaxxinova

"ICEBERG PHENOMENON": practical perspective on correlation of infectious immunosuppression diseases vs Infectious Bronchitis in broiler and layers flocks in Vietnam

GOMES Ludio1, ZUANAZE Marcelo2
1 Vaxxinova International , Regional Office Asia 10/96 The Trendy Building Bangkok ,Thailand
2 Vaxxinova B.V. , Novio Tech Campus, Nijmegen, The Netherlands

Introduction

Infectious bronchitis (IB) is one of the most economically important viral diseases of poultry and can be involved in respiratory disease, drops in egg production, nephritis and enteric problems. At the same time, immunosuppression diseases have a strong impact on IB severity (Naqi, 2001). Among the infectious agents that causes immunosuppression are Marek's Disease Virus (MDV, Calnek et a l., 1998), Reticuloendotheliosis Virus (REV, Walker et al., 1983), Infectious Bursal Disease Virus (IBDV) and Chicken Anemia Virus (CAV, Adair et al., 1991). Routine laboratory tests done in Vietnam, during 2015 and 2016, demonstrated that IBDV and CAV were the most common infectious immunosuppression diseases found in broilers, whereas MDV in commercial layers (personal information).

This practical field study was conducted in order to assess IB outbreaks incidence in broilers (B) and commercial layers (CL) flocks, raised in Northern Vietnam, affected exclusively/simultaneously with IBDV, MDV, REV and CAV.

Materials and methods

Within a period of two years a total of 388 poultry farms(B & CL) were monitored in northern Vietnam. From this amount, 251 flocks were diagnosed as IB outbreak (136 B & 115 in CL) by using macroscopic lesions, RTqPCR and serology (ELISA, Idexx). Broiler flocks were affected within 3 to 5 weeks of age and from 28 to 33 weeks of age for commercial layers. Molecular characterization of IBV, based on classification proposed by Valastro et al. (2016), demonstrated that approximately 74% of cases were positive to genotype I, lineage 16 (I-16), closely related to Q1 TYPE , whereas 21% and 5% to QX and 793-B TYPE respectively. For this work it was selected flocks exclusively IBV diagnosed, thus excluding concomitant outbreaks related to Avian Influenza (AI), Infectious Laryngotracheitis (ILT), Newcastle Disease (ND) and avian Metapneumovirus (aMPV). IBV Vaccination program were very variable receiving from 2 (B) to 8 (CL) doses of live vaccines (793-B and/or Mass) and 1-3 (CL) doses of inactivated vaccines. IBDV vaccination ranged from one single HVT-IBD vector or Immuno-complex vaccination at day-of-age (DOA) to 2 (B) to 4 (CL) doses of live intermediate-plus vaccine in the field. All CL received 1 to 2 doses of Rispens + HVT @ DOC. Approximately 45% of broilers originated from breeders CAV vaccinated.

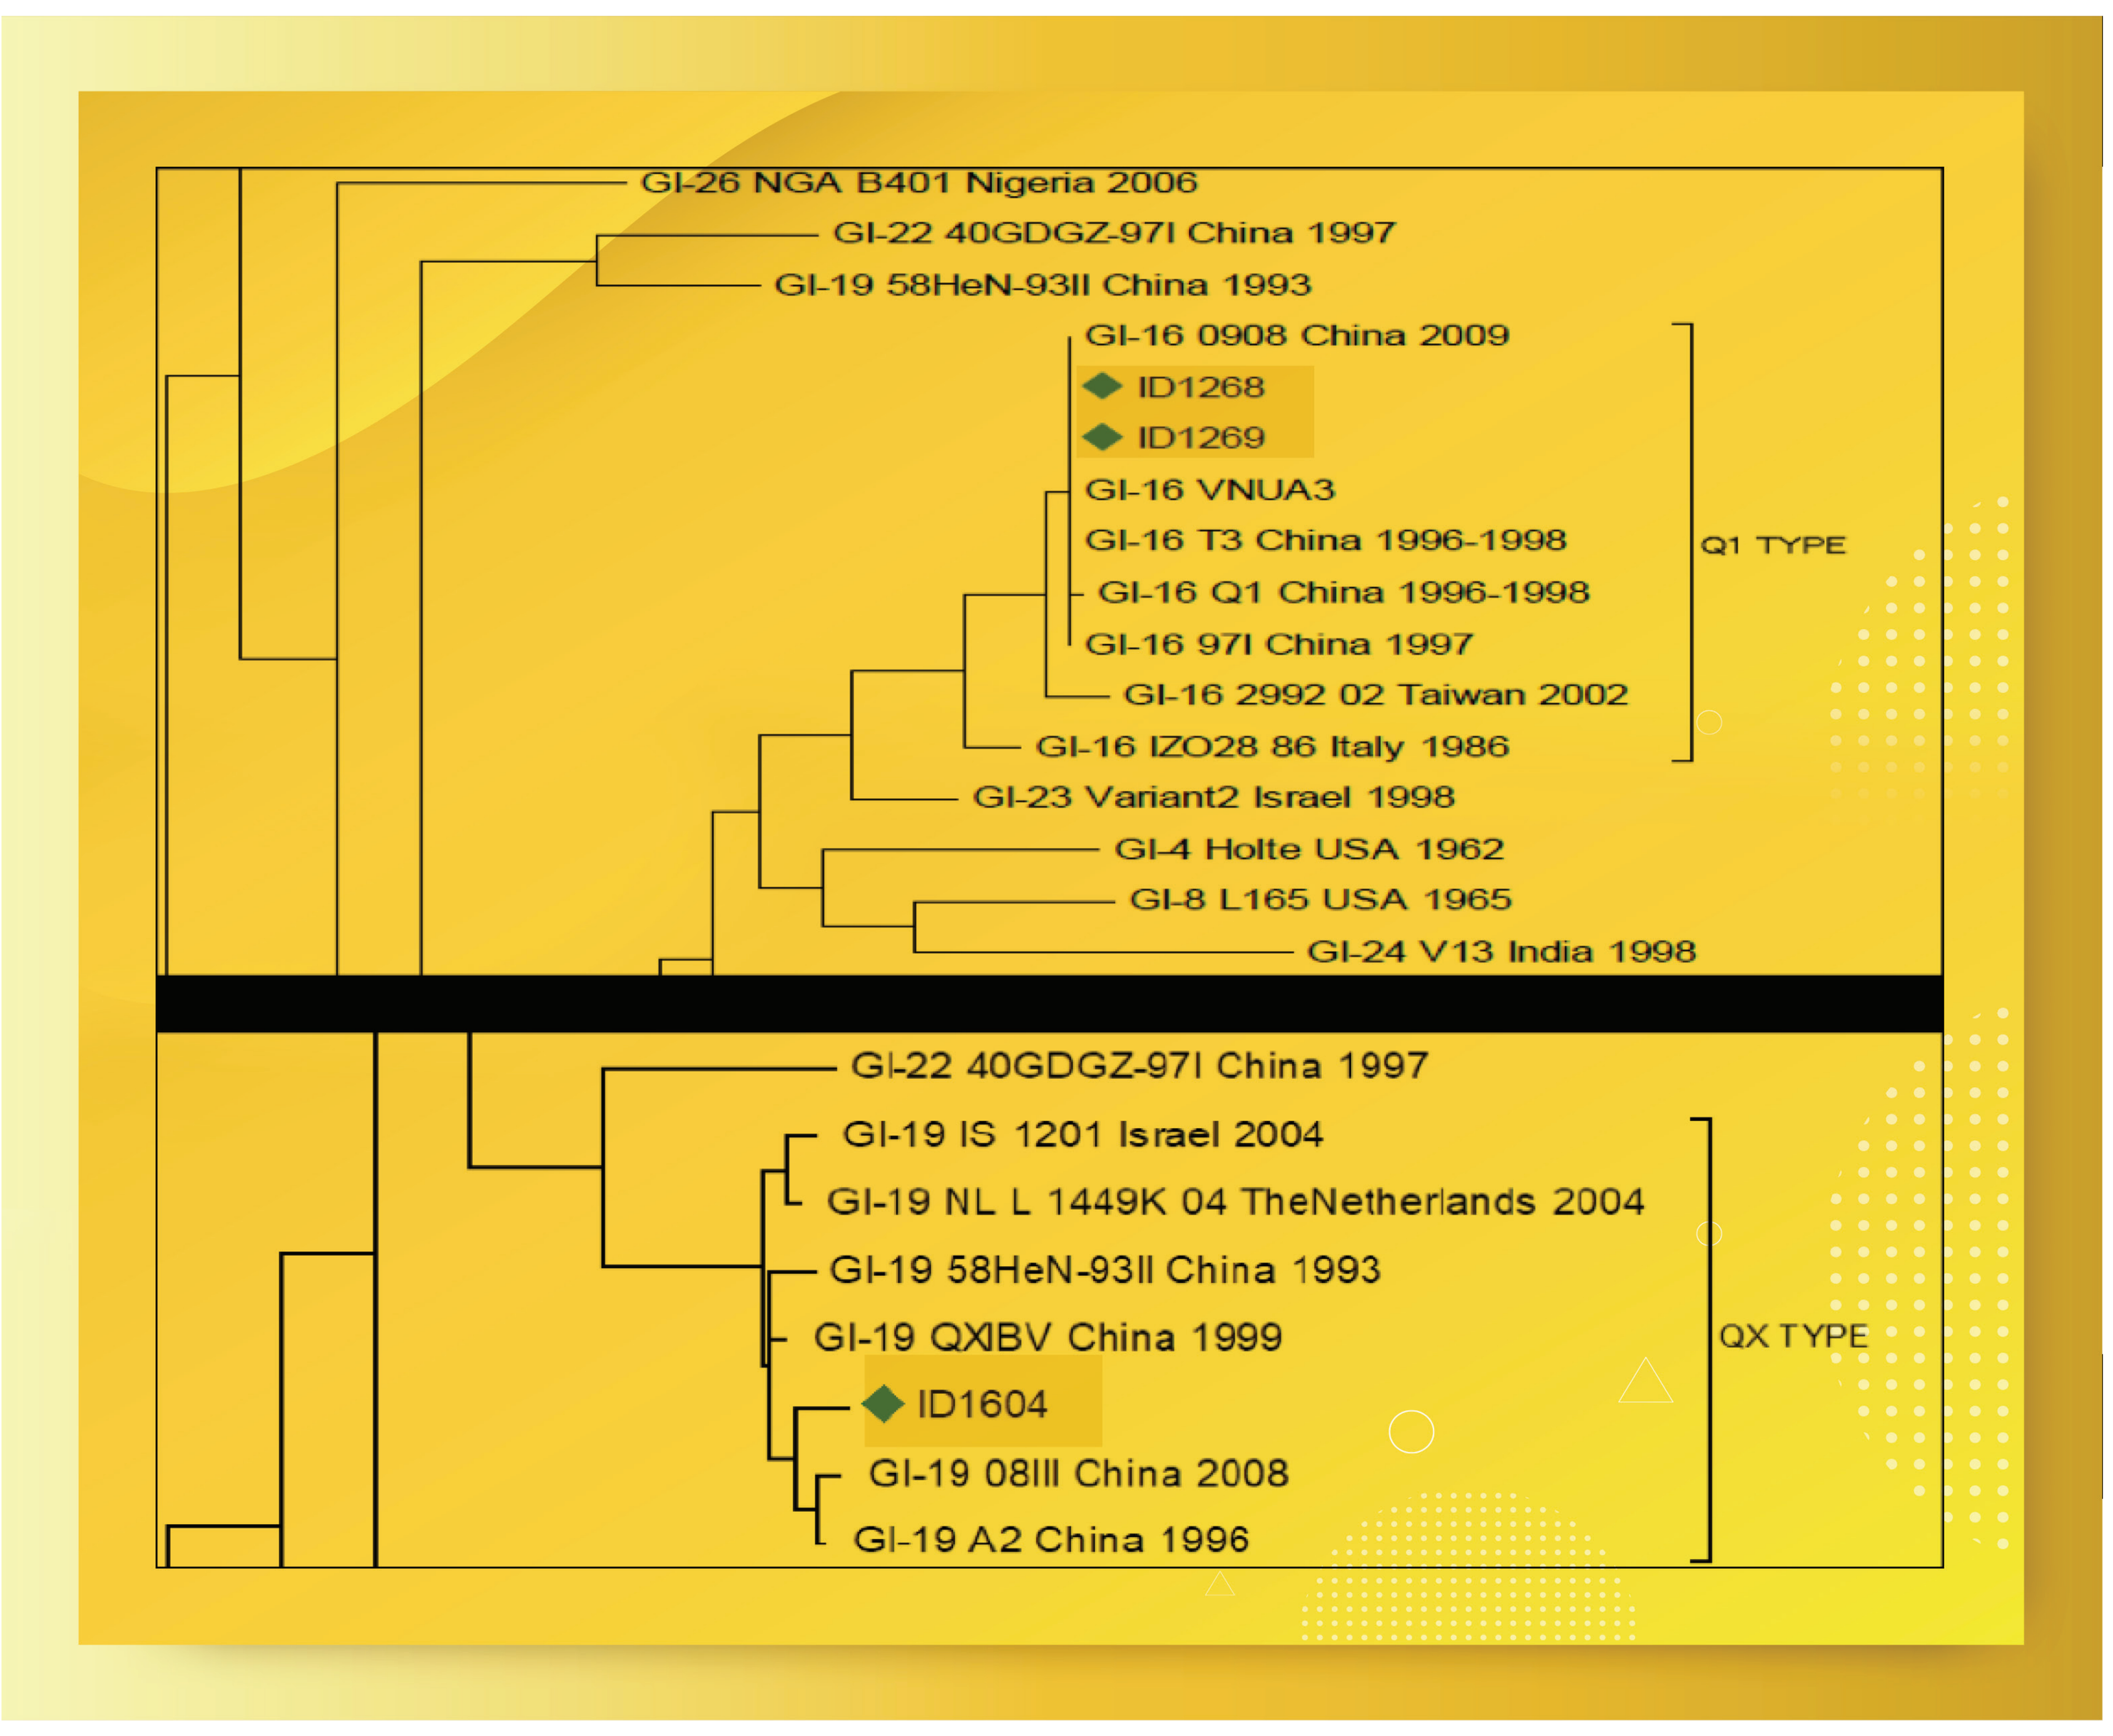
GI-26 NGA B401 Nigeria 2006
GI-22 40GDGZ-97I China 1997
GI-19 58HeN-93II China 1993
GI-16 0908 China 2009
ID1268
ID1269
GI-16 VNUA3
GI-16 T3 China 1996-1998
GI-16 Q1 China 1996-1998
GI-16 97I China 1997
GI-16 2992 02 Taiwan 2002
GI-16 IZO28 86 Italy 1986
GI-23 Variant2 Israel 1998
GI-4 Holte USA 1962
GI-8 L165 USA 1965
GI-24 V13 India 1998
Q1 TYPE
GI-22 40GDGZ-97I China 1997
GI-19 IS 1201 Israel 2004
GI-19 NL L 1449K 04 TheNetherlands 2004
GI-19 58HeN-93II China 1993
GI-19 QXIBV China 1999
ID1604
GI-19 08III China 2008
GI-19 A2 China 1996
QX TYPE

Fig.1. Phylogenetic Analysis of IBV outbreaks based upon nucleic acid composition deriving from IBV S1 sequencing. Roughly 74% of samples (ID1268 and ID1269) diagnosed belongs to group Q1 type group and 21% to QX type (ID1604)

Infectious immunosuppression diseases diagnose

Broilers were monitored by using molecular analysis (IBD RT-PCR & CAV PCR), serology (IBDV & CAV ELISA, Idexx) and macroscopic lesions (≥ 5 birds/necropsy) at 3 and 5 weeks of age. Flocks were considered positive to IBDV when found the combination of at least 2 analyses as with: 1) macroscopic vvIBDV bursa of Fabricius lesions; 2) bursa of Fabricius RT-PCR positiveness to field virus at 3 and/or 5 weeks of age and 3) significant increase in serology titers (Gmean, - ELISA, Idexx). Flocks were considered positive to CAV when found the combination of at least 2 analyses : 1) Thymus hypoplasia / atrophy and/or pale bone marrow; 2) thymus PCR positiveness to field virus at 3 and/or 5 weeks of age and 3) significant increase in serology titers (Gmean - ELISA, Idexx).

Commercial Layers were monitored through PCR and macroscopic lesion at 30 weeks of age. Flocks were considered positive to MDV and/or REV when found the following diagnose combination:1) macroscopic tumors lesions (Fig.02 and 03) and 2) liver and spleen PCR positiveness to field MDV and REV virus.

Results

As shown in Table 1 and 2, a total of 91% (124/136) of broilers and 64% (74/115) of layers flocks were positive to, at least, one of infectious immunosuppression diseases evaluated. Roughly 34% (46/136), 17% (24/136) and 40% (54/136) of broilers were positive to IBD, CAV and combination IBD+CAV respectively. Whilst 32% (37/115), 9% (10/115) and 23% (27/115) were positive to MDV, REV and MDV+REV respectively.

Broilers				
IBD Positive(%)	CAV Positive (%)	IBD + CAV Positive (%)	Negative (%)	Total
33.8 (46/136)	17.6 (24/136)	39.7 (54/136)	8.8 (12/136)	100 (136/136)
Commercial Layers				
MDV Positive(%)	REV Positive (%)	MDV + REV Positive (%)	Negative (%)	Total
32.2 (37/115)	8.7 (10/115)	23.4 (27/115)	35.6 (41/115)	100 (115/115)

Table 1 & 2 . Percentage of positiveness to IBD, CAV and combination IBD+CAV to 136 broilers flocks and MDV , REV and combination MDV + REV to 115 layers flocks diagnosed as IB outbreak. For better understanding of positiveness check material and methods.

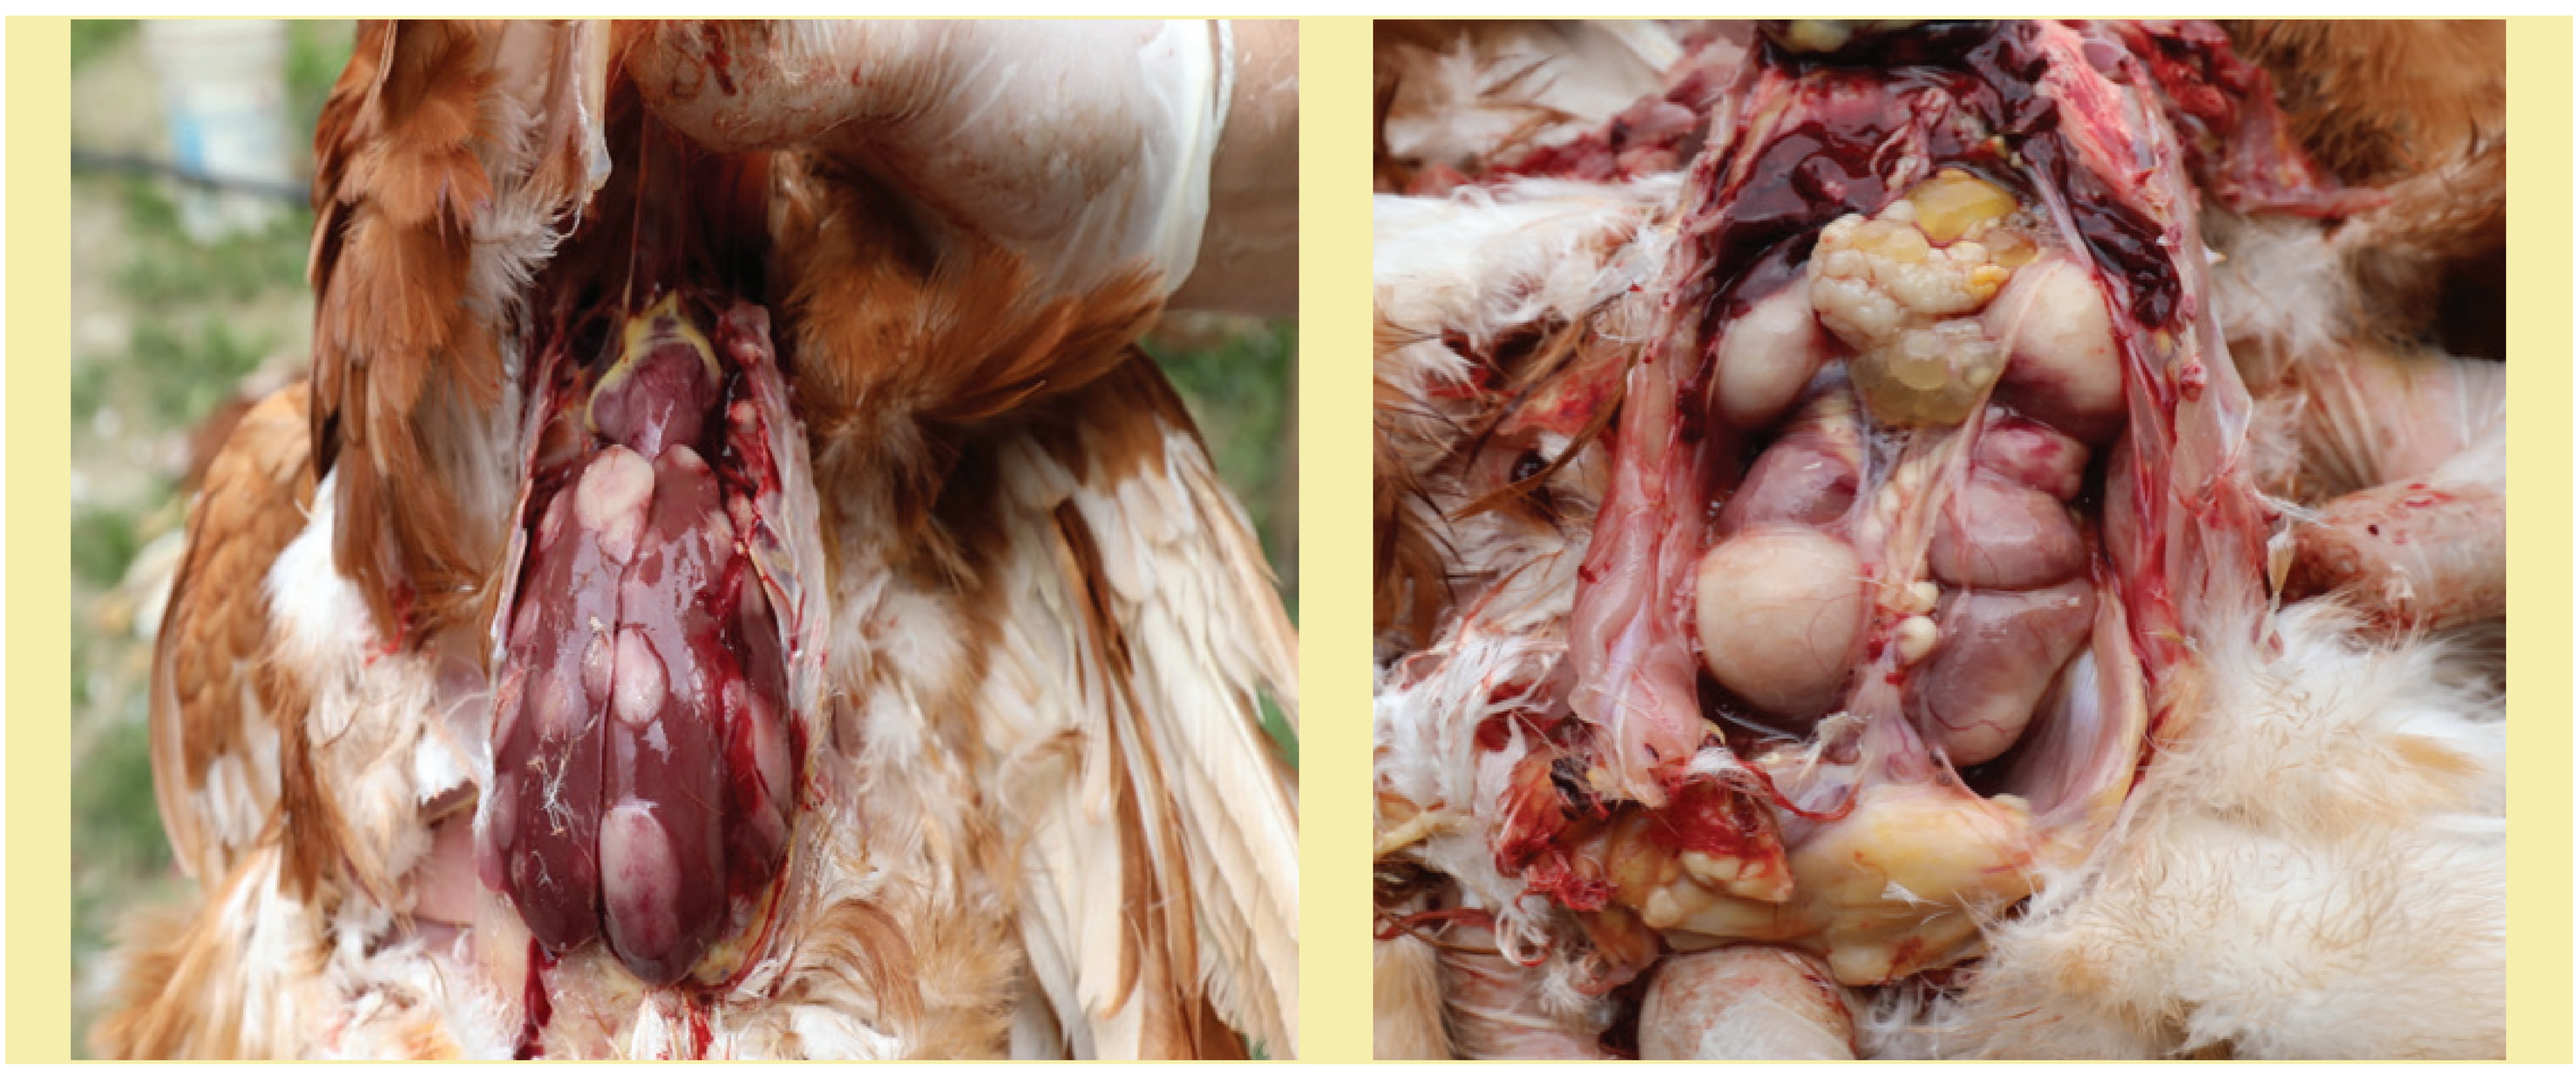

Fig. 2 and 3. Liver tumors (left) and kidneys tumors (right) positive to MDV / REV PCR positive.

DISCUSSION AND CONCLUSION

A set of studies (1,2,4,7 and 8) have established that IBD, CAV, MDV and REV are effective immunosuppressive agents – mainly in young chickens. The effects of immunosuppression may be manifested in a reduction in antibody levels, affecting local a systemic immune response, as well as increased susceptibility to challenge with various agents, included IBV virus. This practical study, done in a low-biosecurity area in Northern Vietnam, demonstrated that 78.8% (198/251) IBV outbreaks in commercial layers and broilers were affected previously or simultaneously by at least one of the immunosuppressive agents evaluated (IBV, CAV, MDV and REV). Interactions between IBDV and CAV might enhance the immunosuppressive damage in broilers and likely it will be the same for MDV and REV for commercial layers. This data reinforce the importance of poultry immune system integrity in relation to IBV control.

References

- **1.Naqi, S, G. Thompson, B Bauman, and H Mohammed. The exacerbating effect of infectious bronchitis virus infection on the infectious bursal disease virus-induced suppression of opsonization by Escherichia coli antibody in chickens. Avian Dis 45:52-60, 2001**
- **2.Calnek, B.W., Harris, R.W., Buscaglia, C., Schat, K.A. and Lucio, B. (1998). Avian Diseases,42, 124-132.**
- **3.Walker, M.H., Rup, B.J., Rubin, A.S. and Bose, H.R., Jr. (1983). Infection and Immunity, 40,225-235.**
- **4.Adair, B.M., Mcneilly, F., Mcconnell, C.D., Todd, D., Nelson, R.T. and Mcnulty, M.S. (1991). Avian Diseases, 35, 783-792.**
- **5.Valastro et al. (Infection, Genetics and Evolution 39:349–364, 2016)**
- **6.Valastro et.al..S1 gene-based phylogeny of infectious bronchitis virus: An attempt to harmonize virus classification. Infection, Genetics and Evolution 39 (2016) 349–364,2016**
- **7.Cloud SS, JK Rosenberger, and HS Lillehoj. Immune dysfunction following infection with chicken anemia agent and infectious bursal disease virus II. Alterations in in vitro lymphoproliferation and in vivo immune responses. Vet Immunol Immunopathol 34:353-366, 1992.**
- **8.F.J. Hoerr, S.B. Lockaby, T.F. Kelly, F. Van Sambeek, J. Cline4, and L. Li. RELATIONSHIP BETWEEN IMMUNOSUPPRESSION AND RESPIRATORY DISEASE, Alabama Department of Agriculture and Industries**
- **Veterinary Diagnostic Laboratories Auburn**